Petosemtamab (MCLA-158) monotherapy or with chemotherapy in metastatic colorectal cancer: Preliminary antitumor activity and safety data from a phase 2 trial

Moh'd Khushman[1], Ruth Vera[2], Thibault Mazard[3], S. DiSean Kendall[4], Elena Élez[5], Francesc Salvà[5], Elisa Fontana[6], Gregory P. Botta[7], Marcos Melián[8], Leon Pappas[9], Ludovic Evesque[10], Anastasia Murat[11], Chris Yan[11], Marina Magin Ferrer[11], Gianluca Laus[11], Fabian Zohren[11], Josep Tabernero[5]

[1]Washington University School of Medicine, St. Louis, MO, USA; [2]Hospital Universitario de Navarra, Pamplona, Spain; [3]Institut Régional du Cancer de Montpellier, Montpellier, France; [4]UCS Cancer Center, Salt Lake City, UT, USA; [5]Vall d'Hebron Instituto de Oncología, Barcelona, Spain; [6]Sarah Cannon Research Institute, London, UK; [7]University of California, San Diego School of Medicine, San Diego, CA, USA; [8]Fundación Instituto Valenciano de Oncología, Valencia Spain; [9]Massachusetts General Hospital, Boston, MA, USA; [10]Centre Antoine Lacassagne, Nice, France; [11]Merus N.V., Utrecht, Netherlands

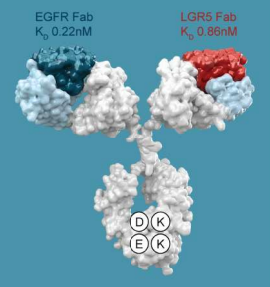
EGFR Fab
K_D 0.22nM
LGR5 Fab
K_D 0.86nM
D K
E K

BACKGROUND

- Anti-EGFR monoclonal antibodies in combination with chemotherapy and as monotherapy are standard of care treatment for metastatic colorectal cancer (mCRC)[1,2]
- LGR5 is expressed in CRC stem-like cells that drive tumor initiation, growth and metastasis[3,4]
- Growth of CRC stem-like cells is dependent on expression of both EGFR and LGR5[3,4]
- Inhibition of EGFR on CRC cells leads to upregulation of LGR5[5,6]
- Petosemtamab: Biclonics® bispecific antibody targeting EGFR and LGR5 that inhibits tumor growth and metastasis[7,8]
- Petosemtamab mechanism of action:[6-8]
 - Inhibition of EGFR ligand binding and downstream signaling
 - Degradation of EGFR via LGR5 internalization in EGFR/LGR5-expressing cells
 - Immune system activation via enhanced ADCC
- Granted two FDA Breakthrough Therapy designations:[9]
 - As monotherapy for 2L+ r/m HNSCC
 - In combination with pembrolizumab for 1L PD-L1+ r/m HNSCC

TRIAL DESIGN AND OBJECTIVES

Phase 2 trial design (NCT03526835)

Key inclusion criteria

- Metastatic CRC
- Left and/or right sided
- Microsatellite stable
- ECOG PS 0–1
- Measurable disease by RECIST v1.1
- 1/2L: EGFRi naïve, RAS/RAF WT by local tissue NGS
- 3L+: EGFRi-, VEGFRi-, and SOC chemotherapy pretreated[a]; KRAS, NRAS, BRAF WT, EGFR ectodomain WT, no ERBB2/HER2 amplification by baseline central ctDNA NGS

1L: Petosemtamab 1500 mg IV + FOLFOX[b] or FOLFIRI[c], Q2W; 28-day cycle (planned enrollment N=40)

2L: Petosemtamab 1500 mg IV + FOLFOX[b] or FOLFIRI[c], Q2W; 28-day cycle (planned enrollment N=40)

3L+: Petosemtamab monotherapy 1500 mg IV, Q2W; 28-day cycle (planned enrollment N=60)

Follow-up

- Tumor assessments Q8W
- Survival follow-up up to 18 months

Objectives

Primary endpoint

ORR using RECIST v1.1 per investigator

Secondary and exploratory endpoints

DOR, PFS, OS, safety, tolerability, and PK characterization

Enrollment and analysis population

Data cutoff date

July 29, 2025

Enrollment

1L combo: 14 patients

2L combo: 14 patients

3L+ monotherapy: 26 patients

Efficacy evaluable population

Patients with ≥1 dose of petosemtamab who had opportunity for ≥8 weeks follow up and ≥1 post baseline tumor assessment or discontinued petosemtamab early due to disease progression, symptomatic deterioration and/or death

1L combo: 10 patients; 4 patients excluded[d]

2L combo: 13 patients; 1 patient excluded[d]

3L+ monotherapy: 20 patients; 6 patients excluded[d]

[a]Oxaliplatin-, Irinotecan-, 5-FU-pretreated; [b]FOLFOX: oxaliplatin 85 mg/m² IV 120 min, leucovorin 400 mg/m² IV 90-120 min, 5-FU bolus 400 mg/m² IV 5 min, and 5-FU 2400 mg/m² IV 46 h; [c]FOLFIRI: irinotecan 180 mg/m² IV 90-120 min, leucovorin 400 mg/m² IV 90-120 min, 5-FU bolus 400 mg/m² IV 5 min, and 5-FU 2400 mg/m² IV 46 h; [d]Reason for exclusion were no post-baseline tumor assessments due to insufficient follow up, unrelated AE or consent withdrawal.

1L, first line; 2L, second line; 3L+, third line and beyond; ADCC, antibody-dependent cellular toxicity; AE, adverse event; C1D1, cycle 1 day 1; CI, confidence interval; CR, complete response; ctDNA, circulating tumor DNA; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; EGFRi, EGFR inhibitor; FDA, Food and Drug Administration; h, hour; HNSCC, head and neck squamous cell carcinoma; IRR, infusion-related reaction; IV, intravenous; K, potassium; LGR5, leucine-rich repeat-containing G-protein coupled receptor; mCRC, metastatic colorectal cancer; Mg, magnesium; NA, not available; NGS, next generation sequencing; ORR, overall response rate; OS, overall survival; PD-L1+, programmed cell death-ligand 1 positive; PFS, progression-free survival; PK, pharmacokinetics; PPE, palmar-plantar erythrodysesthesia; PR, partial response; Q2W, every 2 weeks; Q8W, every 8 weeks; r/m, recurrent/metastatic; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SD, stable disease; SOC, standard of care; TEAE, treatment-emergent adverse event; uCR, unconfirmed complete response; uPR, unconfirmed partial response; VEGFRi, vascular endothelial growth factor receptor inhibitor; WT, wild type.

PATIENT POPULATION

Demographics and disease characteristics	1L combo (N=14)	2L combo (N=14)	3L+ monotherapy (N=26)
Age, years, median (range)	49 (35–73)	55.5 (43–76)	57.5 (30–80)
Male / female, n (%)	8 (57) / 6 (43)	8 (57) / 6 (43)	18 (69) / 8 (31)
ECOG PS, n (%)			
0	7 (50)	4 (29)	12 (46)
1	7 (50)	10 (71)	14 (54)
Race, n (%)			
White	9 (64)	9 (64)	20 (77)
Asian	0	2 (14)	0
Other/not reported	5 (36)	3 (21)	6 (23)
FOLFOX / FOLFIRI, n (%)	10 (71) / 4 (29)	2 (14) / 12 (86)	
Primary site of cancer, n (%)			
Left colon	12 (86)	11 (79)	23 (89)
Right colon	2 (14)	3 (21)	3 (12)
Liver metastases, n (%)	11 (79)	7 (50)	21 (81)
Prior lines of therapy, median (range)	0 (0–0)	1 (1–1)	3 (2-6)

Patient disposition	1L combo (N=14)	2L combo (N=14)	3L+ monotherapy (N=26)
Treatment ongoing, n (%)	13 (93)	11 (79)	11 (42)
Treatment discontinuation, n (%)			
Disease progression	0	2 (14)	13 (50)
Symptomatic deterioration	0	1 (7)	0
Withdrawal by subject	1 (7)	0	0
Study drug-related adverse event	0	0	1 (4)
Unrelated adverse event	0	0	1 (4)
Duration of follow-up, months median (range)	2.8 (0.9–7.1)	4.8 (0.6–12.0)	4.2 (0.4–7.6)

SAFETY

Most common TEAEs irrespective of causality

TEAEs (≥20%), n (%) Preferred Term	Petosemtamab + FOLFOX (N=12) All grades	Petosemtamab + FOLFOX (N=12) Grades 3-5	Petosemtamab + FOLFIRI (N=16) All grades	Petosemtamab + FOLFIRI (N=16) Grades 3-5	Petosemtamab monotherapy (N=26) All grades	Petosemtamab monotherapy (N=26) Grades 3-5
At least 1 TEAE[a]	**12 (100)**	**3 (25)**	**15 (94)**	**11 (69)**	**25 (96)**	**9 (35)**
Acneiform dermatitis	8 (67)	0	5 (31)	0	14 (54)	2 (8)
Nausea	6 (50)	0	5 (31)	0	7 (27)	0
Stomatitis	5 (42)	0	8 (50)	1 (6)	1 (4)	1 (4)
Diarrhea	4 (33)	0	9 (56)	2 (13)	2 (8)	0
Fatigue	4 (33)	0	6 (38)	1 (6)	5 (19)	0
Gastroesophageal reflux disease	4 (33)	0	1 (6)	0	0	0
Constipation	3 (25)	0	5 (31)	0	6 (23)	0
Blood Mg decreased	3 (25)	0	3 (19)	0	5 (19)	1 (4)
Blood K decreased	3 (25)	0	3 (19)	1 (6)	4 (15)	1 (4)
Dyspnea	3 (25)	0	3 (19)	0	3 (12)	0
Vomiting	3 (25)	0	3 (19)	0	4 (15)	0
Dizziness	3 (25)	0	2 (13)	0	3 (12)	0
Embolism	3 (25)	1 (8)	1 (6)	0	0	0
Peripheral sensory neuropathy	3 (25)	0	0	0	0	0
Dry skin	2 (17)	0	4 (25)	0	4 (15)	0
PPE syndrome	1 (8)	0	5 (31)	2 (13)	2 (8)	0
Neutropenia	0	0	5 (31)	3 (19)	0	0
Chills	0	0	5 (31)	0	3 (12)	0
Peripheral edema	0	0	4 (25)	0	0	0

	Petosemtamab + FOLFOX (N=12) All grades	Petosemtamab + FOLFOX (N=12) Grades 3-5	Petosemtamab + FOLFIRI (N=16) All grades	Petosemtamab + FOLFIRI (N=16) Grades 3-5	Petosemtamab monotherapy (N=26) All grades	Petosemtamab monotherapy (N=26) Grades 3-5
IRR[b]	4 (33)	0	7 (44)	0	10 (30)	1 (4)

- Petosemtamab's safety profile in mCRC is consistent with its established safety profile in r/m HNSCC
- No significant overlapping toxicities identified in combination with FOLFOX/FOLFIRI
- No new safety signals identified
- No G5 or G4 treatment-related TEAEs observed in combination with FOLFOX/FOLFIRI
- No G5 treatment-related TEAEs observed with petosemtamab monotherapy
- One patient discontinued petosemtamab monotherapy due to treatment-related G4 hypomagnesemia
- Cutaneous side effect profile[c] appears favorable and manageable without pharmacologic prophylaxis
- IRRs were managed with premedication and prolonged infusion on C1D1; no discontinuations due to IRRs

[a]Most common TEAEs, irrespective of causality, are defined as AEs with onset date on or after date of first administration of study drug and ≤30 days post-treatment; [b]IRR is a composite term for one or multiple signs/symptoms during the 24-hour period after initiating the petosemtamab infusion, judged by investigators as an IRR; [c]Composite term including dermatitis acneiform, folliculitis, rash maculo-papular, skin fissures, PPE syndrome.

EFFICACY

Petosemtamab + FOLFOX/FOLFIRI in 1L mCRC (n=10)

Best percent change in sum of target lesions from baseline

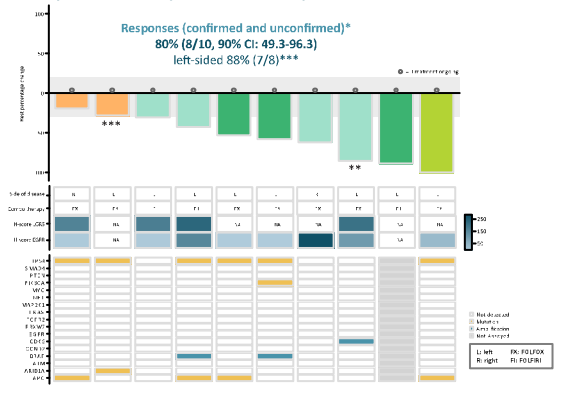
Responses (confirmed and unconfirmed)*
80% (8/10, 90% CI: 49.3-96.3)
left-sided 88% (7/8)***
L: left FX: FOLFOX
R: right FI: FOLFIRI

Time to response and duration of exposure

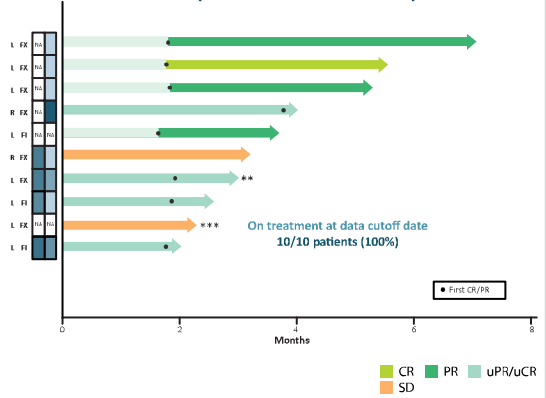
On treatment at data cutoff date
10/10 patients (100%)
First CR/PR
Months
CR
PR
uPR/uCR
SD

*RECIST v1.1, per investigator; **Unconfirmed response was confirmed post data cutoff; ***Unconfirmed PR documented post data cutoff leading to 100% responses left-sided (8/8).

Petosemtamab + FOLFOX/FOLFIRI in 2L mCRC (n=13)

Best percent change in sum of target lesions from baseline[a]

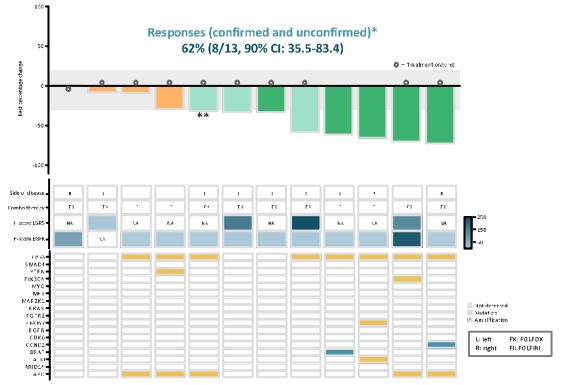
Responses (confirmed and unconfirmed)*
62% (8/13, 90% CI: 35.5-83.4)
L: left FX: FOLFOX
R: right FI: FOLFIRI

Time to response and duration of exposure

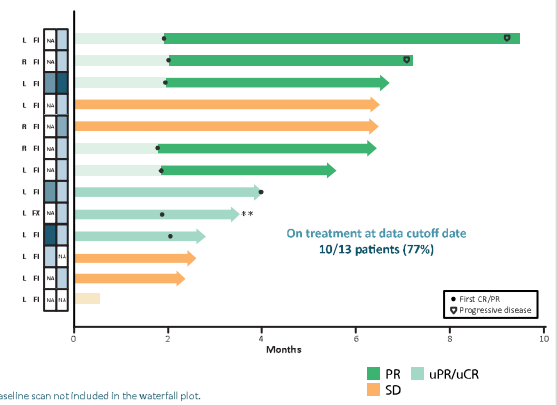
On treatment at data cutoff date
10/13 patients (77%)
First CR/PR
Progressive disease
Months
PR
uPR/uCR
SD

[a]One patient with early symptomatic deterioration and no post-baseline scan not included in the waterfall plot.
*RECIST v1.1, per investigator; **Unconfirmed response was confirmed post data cutoff.

Petosemtamab monotherapy in 3L+ mCRC (n=20)

Best percent change in sum of target lesions from baseline

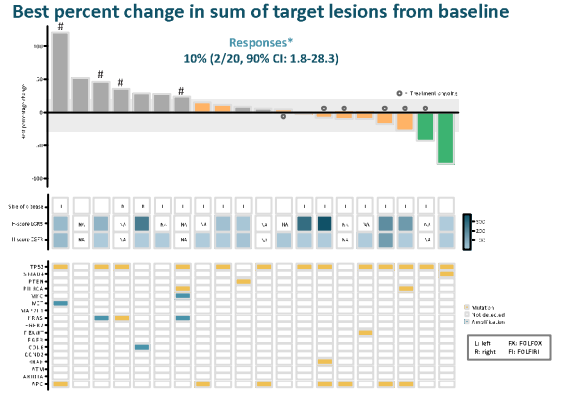
Responses*
10% (2/20, 90% CI: 1.8-28.3)
L: left FX: FOLFOX
R: right FI: FOLFIRI

Time to response and duration of exposure

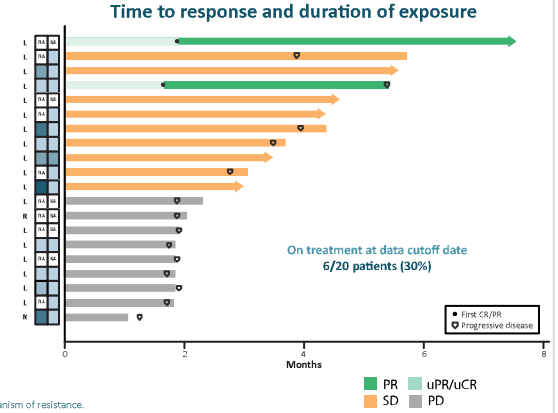
On treatment at data cutoff date
6/20 patients (30%)
First CR/PR
Progressive disease
Months
PR
uPR/uCR
SD
PD

*RECIST v1.1, per investigator; #Identified mechanism of resistance.

Intrahepatic tumor responses with petosemtamab + FOLFOX/FOLFIRI: Case studies

- Among 14 patients with measurable target liver lesions, 10 (71%) showed intrahepatic objective response
 - 1L: 6/8 (75%)
 - 2L: 4/6 (67%)

67-year-old male with metastatic sigmoid adenocarcinoma

- No prior systemic treatment
- Treated with petosemtamab + FOLFOX; unconfirmed PR, confirmed post data cutoff

Baseline | 8 weeks | 16 weeks

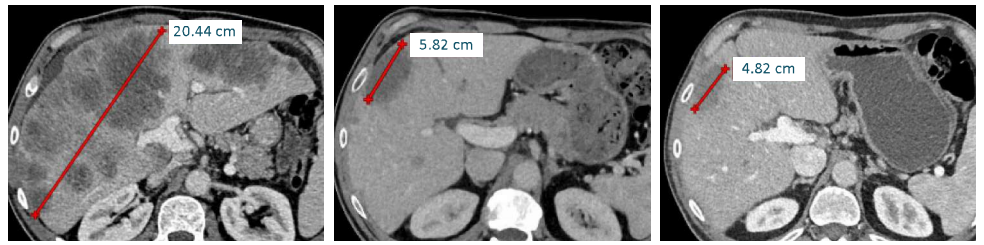
20.44 cm
5.82 cm
4.82 cm

73-year-old male with metastatic rectal adenocarcinoma

- 1 prior line of systemic treatment in the metastatic setting
- Treated with petosemtamab + FOLFIRI; confirmed PR

Baseline | 8 weeks | 16 weeks

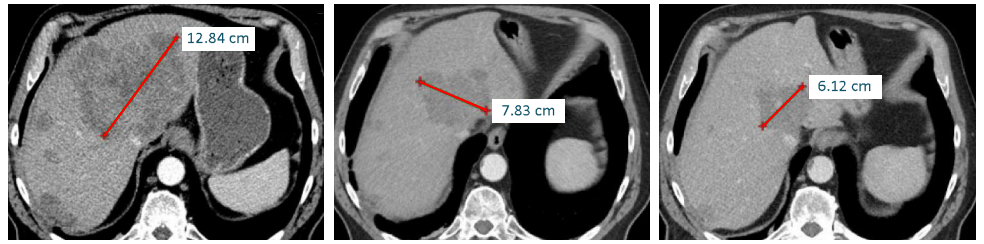
12.84 cm
7.83 cm
6.12 cm

CONCLUSIONS

Petosemtamab in combination with FOLFOX/FOLFIRI and as monotherapy demonstrates promising efficacy and a well-tolerated safety profile in mCRC

Substantial antitumor activity:

- 1L: 8/10 (80%) confirmed + unconfirmed responses and 2 SD with tumor shrinkage continuing treatment
 - Left-side: 7/8 (88%) confirmed + unconfirmed responses
- 2L: 8/13 (62%) confirmed + unconfirmed responses and 3 SD with tumor shrinkage continuing treatment
- 3L+: 2/20 (10%) confirmed responses and 4 SD with tumor shrinkage continuing treatment

Favorable safety profile with no new safety signals identified:

- Cutaneous side effect profile appears favorable and manageable without pharmacologic prophylaxis

Clinical development continues:

- 1L and 2L mCRC in combination with FOLFOX/FOLFIRI and 3L+ mCRC monotherapy enrolling (NCT03526835)
- Phase 3 registrational trials in r/m HNSCC enrolling
 - 1L PD-L1+ with pembrolizumab (LiGeR-HN1, NCT06525220)
 - 2/3L monotherapy (LiGeR-HN2, NCT06496178)

References

1. Douillard JY, et al. *N Engl J Med.* 2013;369(11):1023-1034.
2. Watanabe J, et al. *JAMA.* 2023;329(15):1271-1282.
3. Katoh M, *Int J Oncol.* 2017;51(5):1357–1369.
4. Xu L, et al. *Stem Cell Res Ther.* 2019;10(1):219.
5. Shimokawa M, et al. *Nature.* 2017;545:187-192.
6. Slobodnyuk K, et al. Presented at 2025 AACR-NCI-EORTC international conference [poster B042].
7. Herpers B, et al. *Nat Cancer.* 2022;3(4):418–436.
8. Lundberg AS, et al. *Cancers (Basel).* 2025;17(10):1665.
9. Merus N.V. News release. 2025. Available at https://ir.merus.nl/news-releases/news-release-details/petosemtamab-granted-breakthrough-therapy-designation-us-fda-1l/. Accessed August 4, 2025.

Acknowledgments

We thank the patients and their families, the investigators and their teams, Merus colleagues (including Marina Magin Ferrer); clinical research organizations, and vendors. Study sponsored by Merus N.V.

Presenting author disclosures: M. Khushman is a consultant or advisor for TriSalusLife Sciences, Cardinal Health, BMS, AbbVie, Exelixis, Elevar Therapeutics, Guardant Health, and Merus N.V.; and has received research funding from AbbVie, FOGPharma, IgM Biotherapeutics, Merus N.V., GlaxoSmithKline, and BeiGene.

Presented at AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics, October 22-25, 2025, Hynes Convention Center, Boston, MA.

ClinicalTrials.gov Identifier: NCT03526835

Contact information: Dr. Ernesto Wasserman, E.Wasserman@merus.nl

Copies of this poster obtained through QR (quick response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.